San Antonio Breast Cancer Symposium – December 5-9, 2017

P1-07-14

Real-life Analysis Evaluating >1000 N0/N1mi Estrogen Receptor (ER)+ Breast Cancer Patients for whom Treatment Decisions Incorporated the 21-gene Recurrence Score (RS) Result: Clinical Outcomes with Median Follow up of Approximately 9 Years

Salomon M. Stemmer[1,2], Shulamith Rizel[1], David B. Geffen[3], Mariana Steiner[4], Lior Soussan-Gutman[5], Avital Bareket-Samish[6], Debbie McCullough[7], Christer Svedman[7], Steven Shak,[7] Beatrice Uziely[8], Georgeta Fried[9], Ora Rosengarten[10], Amit Itay[11], Bella Nisenbaum[12], Larisa Ryvo[13], Daniela Katz[14], Margarita Toker[3], Nicky Liebermann[15], Noa Ben-Baruch[16]

[1]Davidoff Center, Rabin Medical Center, Petah Tikva, Israel; [2]Sackler Faculty of Medicine, Tel Aviv University, Tel Aviv, Israel; [3]Department of Oncology, Soroka University Medical Center and the Faculty of Health Sciences, Ben-Gurion University of the Negev, Beer Sheva, Israel; [4]Lin Medical Center, Haifa, Israel; [5]Oncotest Division, Teva Pharmaceutical Industries, Ltd., Shoham, Israel; [6]BioInsight Ltd, Zichron Yaakov, Israel; [7]Genomic Health Inc., Redwood City, CA; [8]Sharett Institute of Oncology, Hadassah-Hebrew University Medical Center, Jerusalem, Israel; [9]Oncology Dept., Rambam Health Care Campus, Haifa, Israel; [10]Oncology Institute, Shaare Zedek Medical Center, Jerusalem, Israel; [11]Sheba Medical Center at Tel HaShomer, Ramat Gan, Israel; [12]Oncology Dept., Meir Medical Center, Kfar Saba, Israel; [13]Oncology Dept., Tel-Aviv Sourasky Medical center, Tel Aviv, Israel; [14]Assaf Harofeh Medical Center, Zerifin, Isarel; [15]Community Division, Clalit Health Services, Tel Aviv, Israel; [16]Oncology Dept., Kaplan Medical Center, Rehovot, Israel.

BACKGROUND

- The 21-gene Oncotype DX® Breast Recurrence Score™ (RS) assay, is a validated prognosticator/predictor of chemotherapy (CT) benefit in estrogen receptor (ER)+ human epidermal growth factor receptor 2 (HER2)-negative early-stage breast cancer (BC).[1-9] Five-year outcome data from over 50,000 patients treated according to the assay has also been reported (Clalit, SEER).[10-12] Based on outcome data from extensive validation studies with long follow-up, the assay has been incorporated into major international guidelines,[13-16] as well as into the recent edition of the American Joint Committee on Cancer (AJCC) BC staging manual,[17] and is widely used to guide treatment decisions.
- ER+ BC patients, have a protracted recurrence risk with approximately half of all distant recurrences occurring after 5 years with a continuum of relapse until 20 years.[18,19]
 - Since 2014, the American Society of Clinical Oncology (ASCO) has recommended extending the duration of endocrine therapy in hormone receptor (HR)+ BC from 5 to 10 years.[20]
- In Israel, Clalit Health Services (CHS), the largest HMO in Israel, started reimbursing the RS assay in 2006. We have previously reported treatment decisions and 5-year clinical outcomes in patients who underwent RS testing through CHS and whose treatment decisions in real-life clinical practice incorporated the RS results.[10,11] Our findings were consistent with the validation studies demonstrating the prognostic value of the assay, and support the use of endocrine therapy alone in N0 patients with RS results ≤25 and in node-positive patients (N1mi, 1-3 positive nodes) with RS results <18.[18,19] Ten-year outcome data from cohorts treated according to the RS results have not yet been presented.

OBJECTIVE

- To characterize 10-year distant recurrence rates in N0/N1mi ER+ HER2-negative BC patients who underwent RS testing through CHS.

METHODS

Study Design and Patient Population

- This retrospective analysis of the prospectively designed CHS registry investigated the relationship between the RS result, adjuvant treatments received, and distant recurrence/survival in patients with ER+ HER2-negative N0/N1mi BC in real-life clinical practice. Collecting outcome data from all CHS RS-tested patients was planned by CHS, in concert with assay reimbursement approval.
 - Inclusion criteria: All CHS patients with ER+ HER2-negative N0/N1mi BC who underwent RS testing between 1/2006 (CHS approval of the assay) and 12/2009 (N0) or 6/2010 (N1mi).
 - Exclusion criteria: ER-negativity by immunohistochemistry (IHC) and reverse transcription polymerase chain reaction (RT-PCR); HER2 positivity by IHC or RT-PCR; adjuvant trastuzumab treatment; neoadjuvant treatment; metastatic disease at/within 6 months of testing; and adjuvant CT for another malignancy within 6 months of testing.
- Endpoints: Kaplan-Meier (KM) estimates for 10-year risk of distant recurrence (primary) and BC death (secondary) by RS risk group. Exploratory endpoints included distant recurrence/BC death analysis by clinicopathological subgroups as well as by CT use.
- The study was approved by the institutional review boards of the CHS Community Division and participating medical centers, and was granted a waiver for obtaining patient consent.

Data Source

- Data sources used: The Teva Pharmaceutical Industries database (for RS results and patient/tumor characteristics); medical records and the CHS claims arm (for treatments received and recurrence/death).

Statistical Analysis

- This analysis is an exploration of the maturing data with long-term follow up.
- Descriptive statistics were used to summarize clinicopathological characteristics and adjuvant CT decisions.
- Log-rank test was used to compare distant recurrence rates and BC deaths across RS groups.
- Hazard ratios and 95% confidence intervals (CIs) were calculated using Cox regression models.

RESULTS

Patient Characteristics

- The final cohort included 1540 N0/N1mi BC patients who underwent RS testing between 1/2006 and 12/2009 (N0) or 6/2010 (N1mi). The overall median follow-up in patients was 8.9 (interquartile range, 5.9-9.8) years; for N0 patients, the median follow-up was 9.0 years and for N1mi patients, it was 7.6 years.

Table 1. Baseline patient and tumor characteristics.

	N = 1540
Female, %	1527 (99%)
Median (interquartile range) age, years	60 (52-66)
Age category, %	
<40 years	37 (2%)
40-49 years	215 (14%)
50-59 years	507 (33%)
60-69 years	536 (35%)
70-79 years	226 (15%)
≥80 years	19 (1%)
Median (interquartile range) tumor size in the greatest dimension, cm	1.5 (1.2-2.0)
Tumor size category, %	
≤1 cm	323 (21%)
>1 - 2 cm	854 (55%)
>2 cm	351 (23%)
Unknown	12 (1%)
Tumor grade category, %	
Grade 1	224 (15%)
Grade 2	774 (50%)
Grade 3	255 (17%)
Not applicable/Unknown[a]	287 (19%)
Histology, %	
IDC	1249 (81%)
ILC	180 (12%)
Papillary	16 (1%)
Mucinous/colloid	42 (3%)
Other/unknown	53 (4%)
Nodal involvement, %	
N0	1365 (89%)
N1mi	175 (11%)

IDC, invasive ductal carcinoma; ILC, invasive lobular carcinoma.

[a]60% of unknown tumor grade are ILC.

- RS distribution was: 50% RS <18 (18% RS <11, 32% RS 11-17), 39% RS 18-30 (30% RS 18-25, 9% RS 26-30), and 10% RS ≥31.
- Adjuvant CT use was <1%, 3%, 17%, 52%, and 89% for RS <11, 11-17, 18-25, 26-30, ≥31, respectively, consistent with the RS result (overall CT use, 20%).

Distant Recurrence Rates and BC Death Rates

- KM estimates for 10-year distant recurrence and BC death rates in both N0 and N1mi patients differed significantly between the RS groups (P <0.001; log-rank test; **Figure 1**).
- The RS result was predictive of late recurrence (P = 0.022; data not shown).

Figure 1. KM distant recurrence and BC specific mortality curves by RS groups.

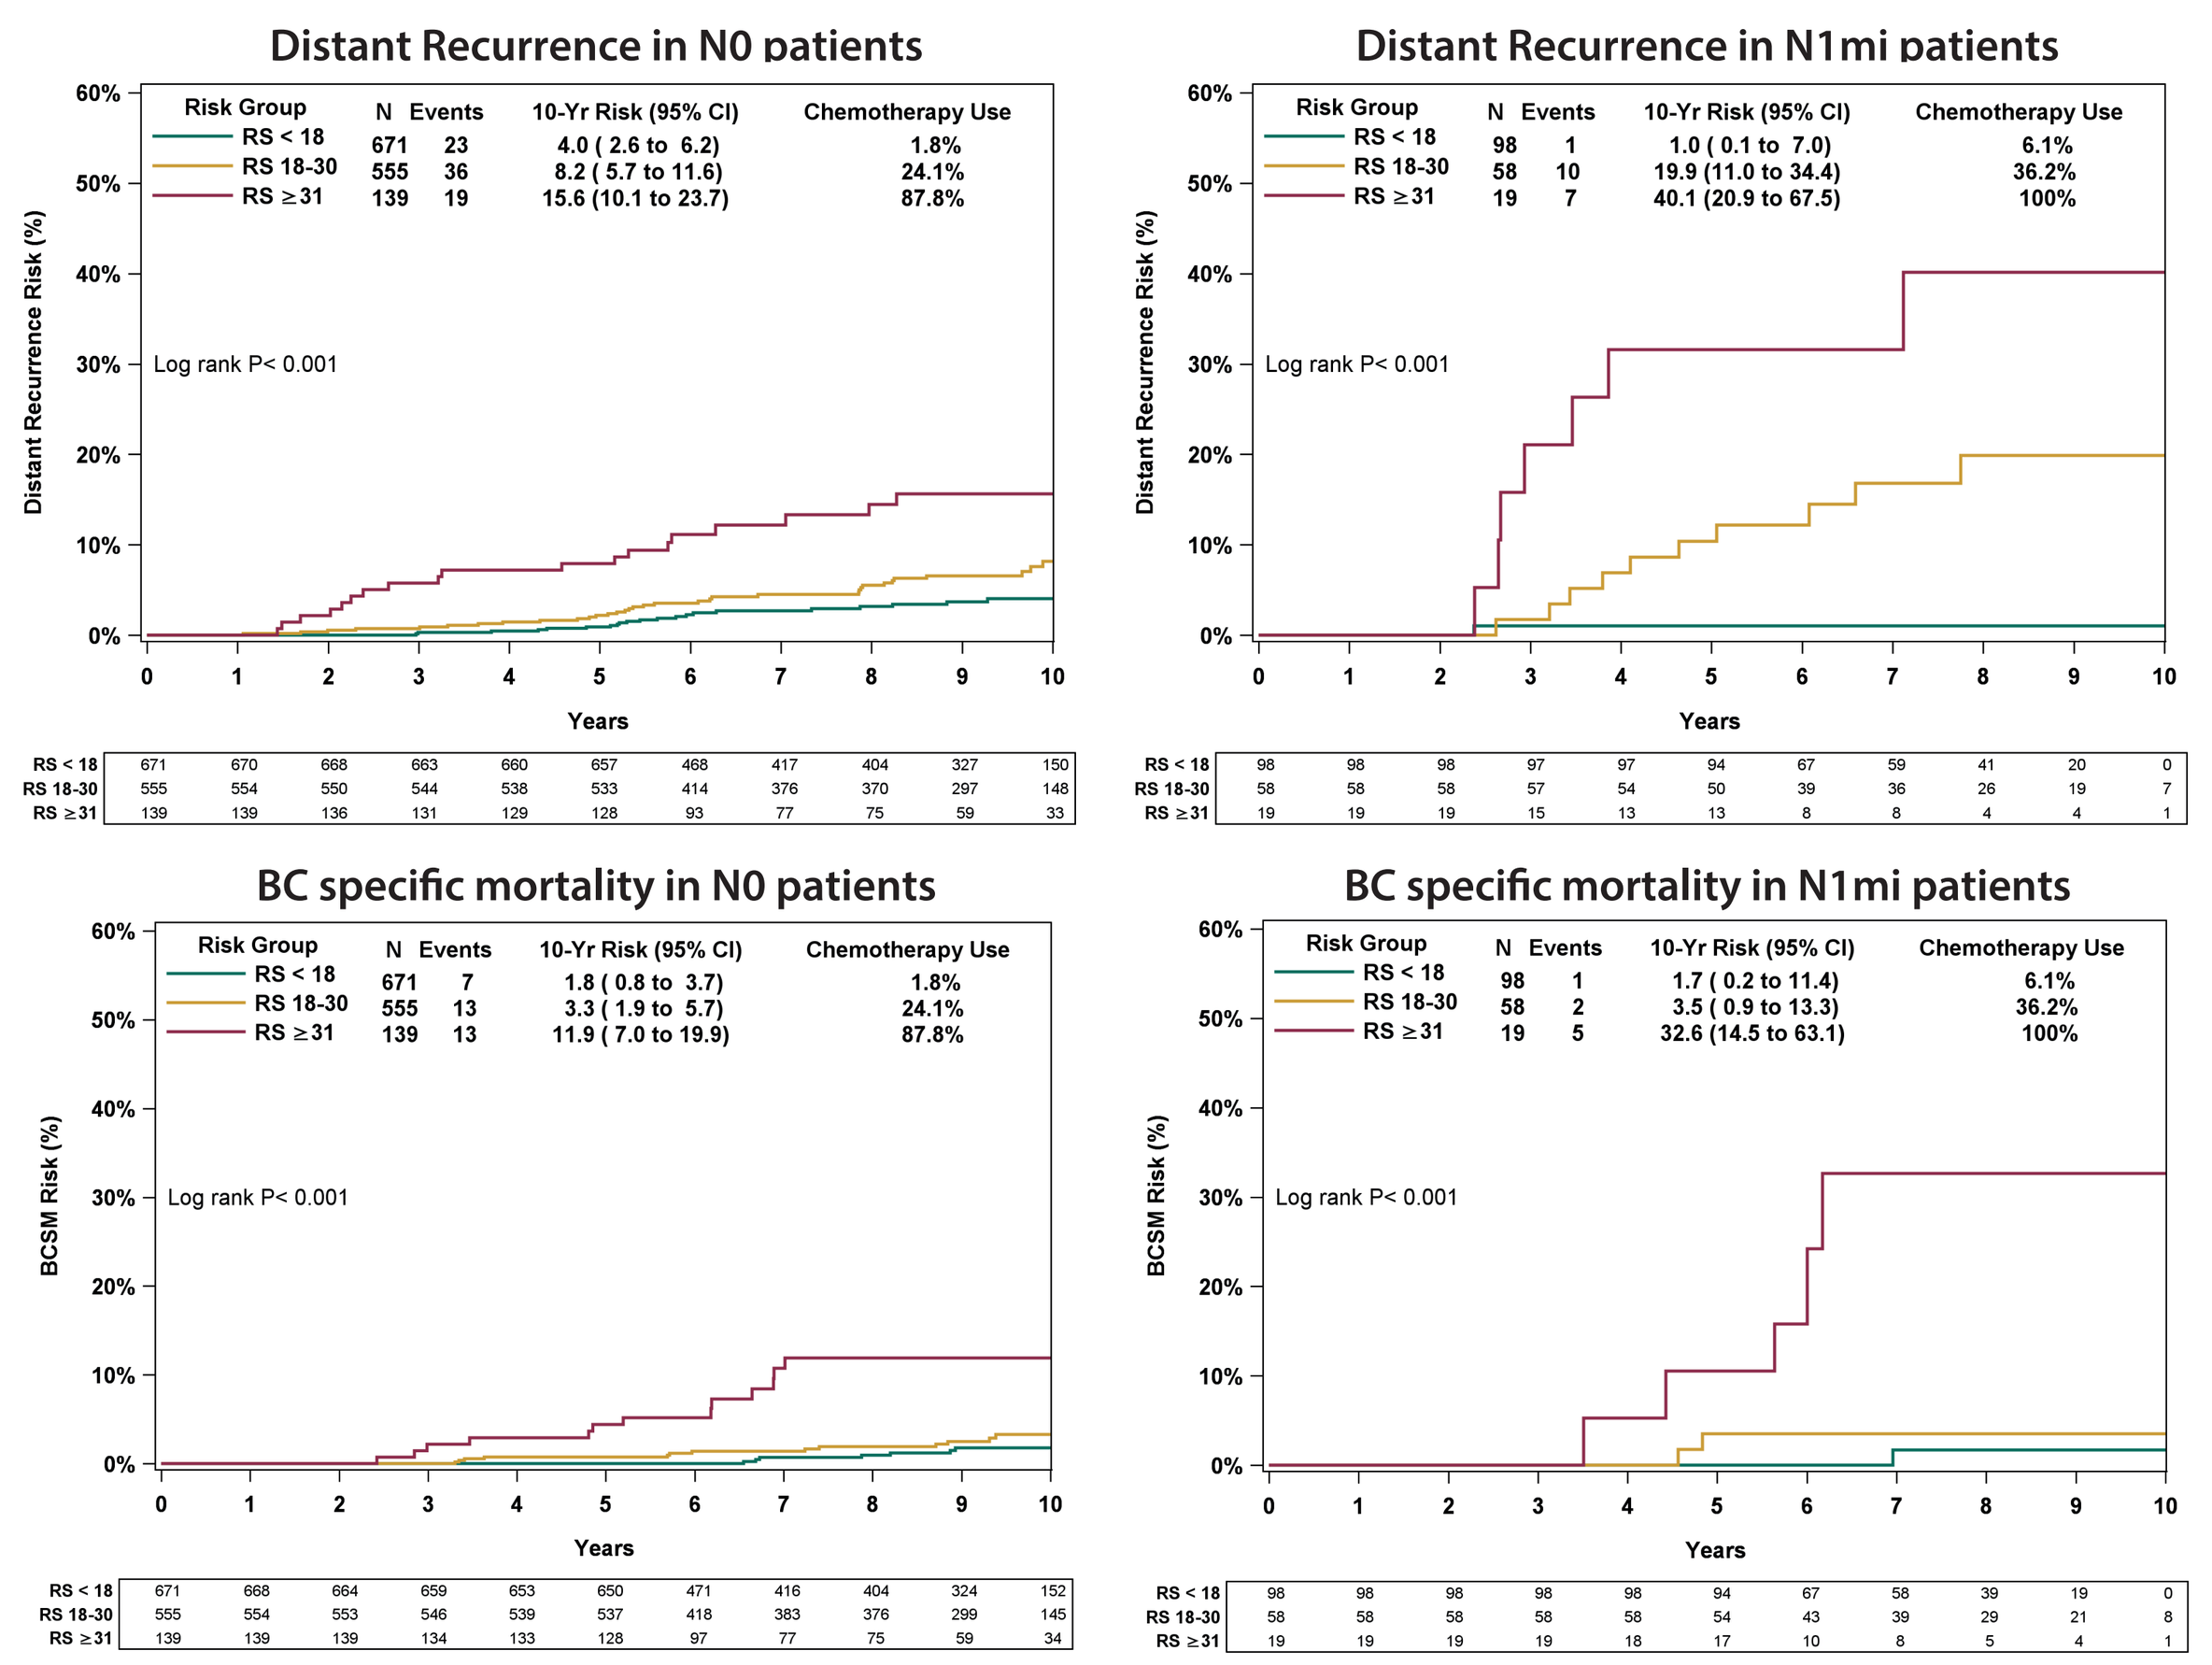
Distant Recurrence in N0 patients
Risk Group N Events 10-Yr Risk (95% CI) Chemotherapy Use
RS < 18 671 23 4.0 (2.6 to 6.2) 1.8%
RS 18-30 555 36 8.2 (5.7 to 11.6) 24.1%
RS ≥31 139 19 15.6 (10.1 to 23.7) 87.8%
Log rank P< 0.001
Distant Recurrence in N1mi patients
RS < 18 98 1 1.0 (0.1 to 7.0) 6.1%
RS 18-30 58 10 19.9 (11.0 to 34.4) 36.2%
RS ≥31 19 7 40.1 (20.9 to 67.5) 100%
Log rank P< 0.001
BC specific mortality in N0 patients
RS < 18 671 7 1.8 (0.8 to 3.7) 1.8%
RS 18-30 555 13 3.3 (1.9 to 5.7) 24.1%
RS ≥31 139 13 11.9 (7.0 to 19.9) 87.8%
Log rank P< 0.001
BC specific mortality in N1mi patients
RS < 18 98 1 1.7 (0.2 to 11.4) 6.1%
RS 18-30 58 2 3.5 (0.9 to 13.3) 36.2%
RS ≥31 19 5 32.6 (14.5 to 63.1) 100%
Log rank P< 0.001
Years

The box under each graph presents the number of patients at risk at each time point.
Two-degree of freedom (df) log-rank P values were calculated from all data.

Risk of Distant Recurrence/BC Death in N0 Patients Treated with Endocrine Therapy Alone

- We analyzed the 94% of RS<18 patients and the 73% of RS 18-30 patients who received endocrine therapy alone, and found low distant recurrence risk/BC death rates in the RS<18 patients (**Figure 2**).

Figure 2. KM distant recurrence and BC specific mortality curves in N0 patients with RS<18 and RS 18-30 who received endocrine therapy alone.

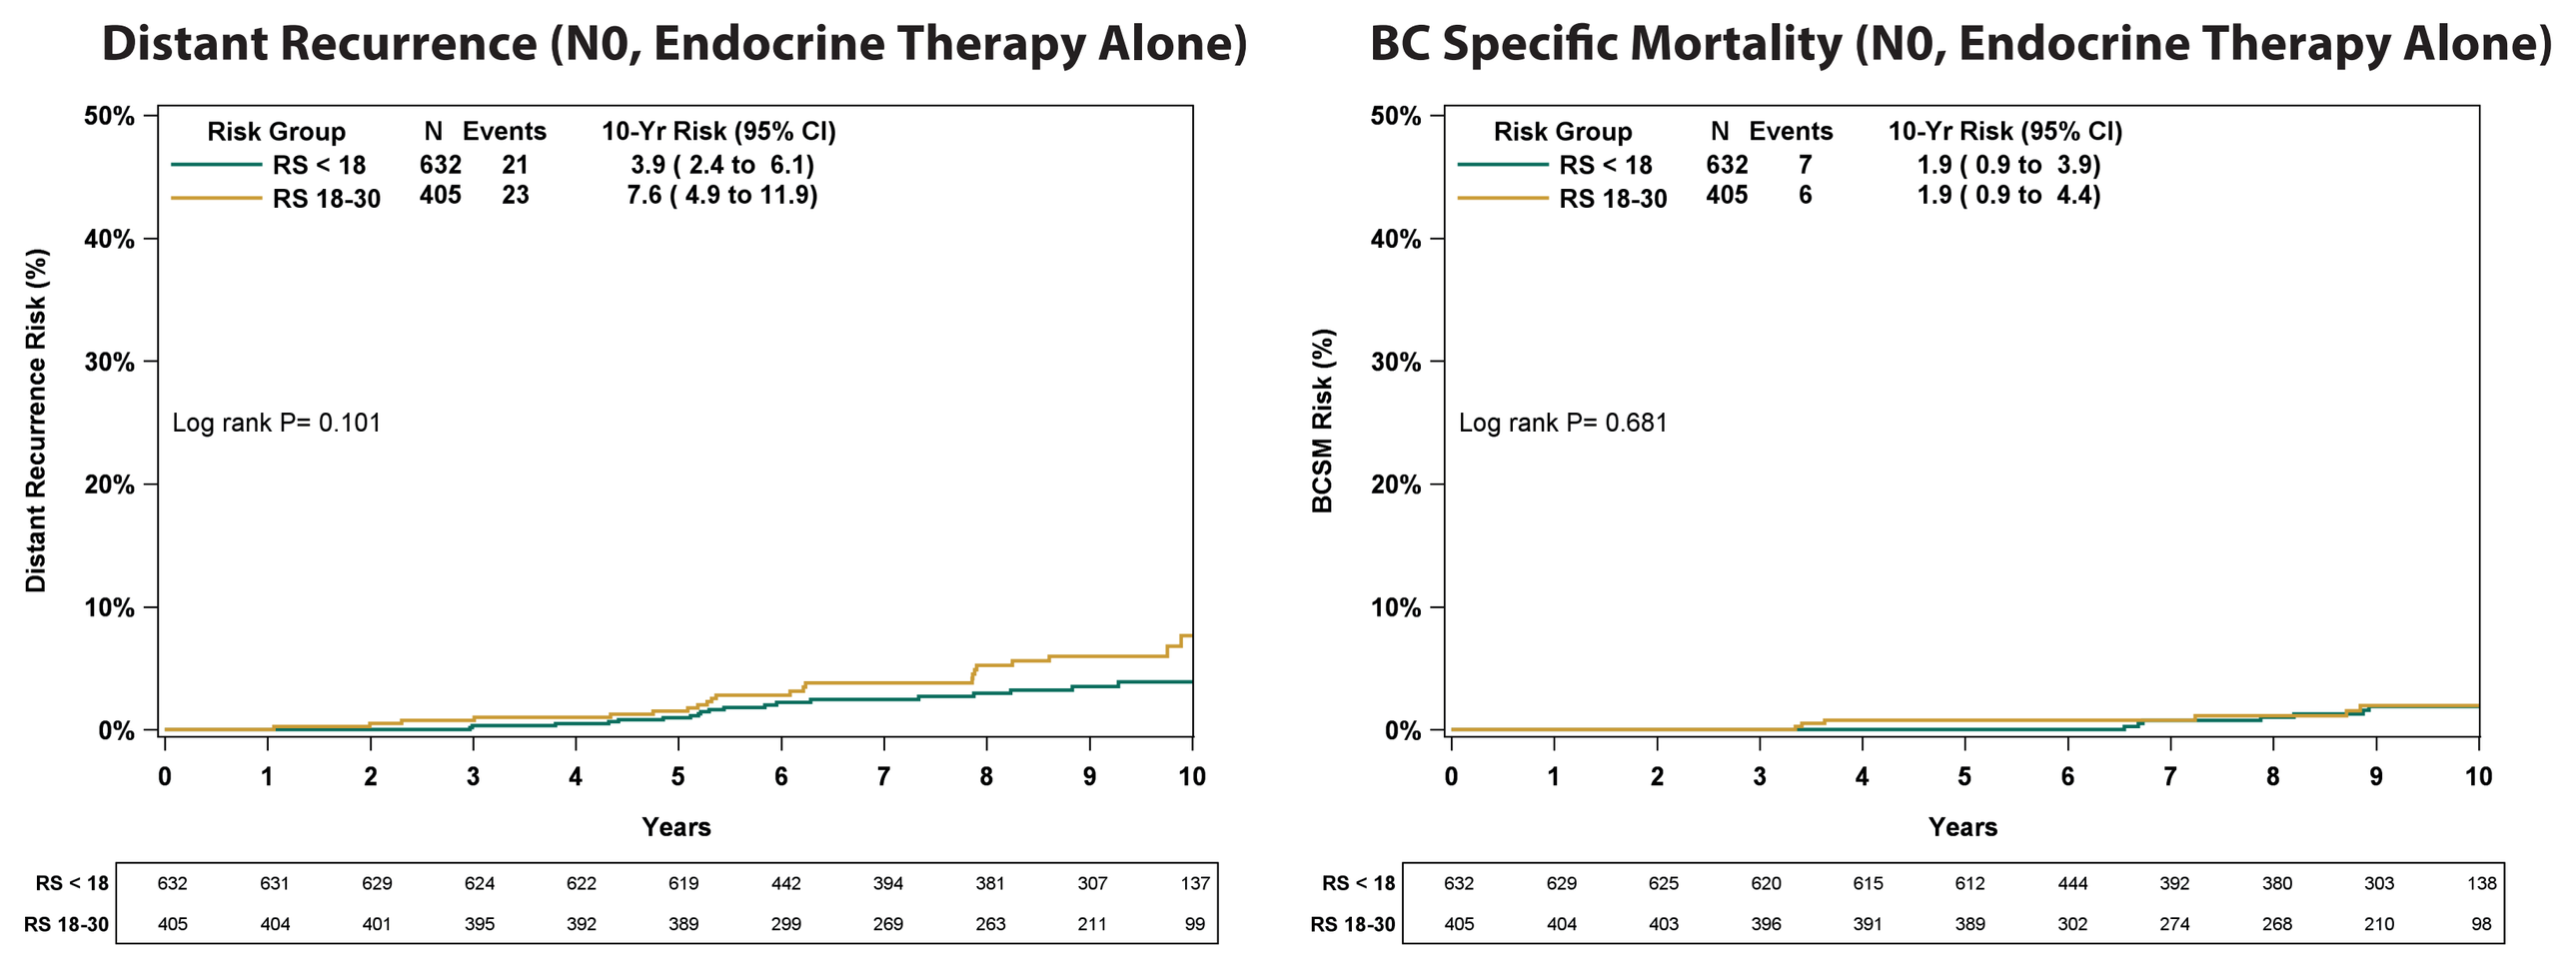
Distant Recurrence (N0, Endocrine Therapy Alone)
Risk Group N Events 10-Yr Risk (95% CI)
RS < 18 632 21 3.9 (2.4 to 6.1)
RS 18-30 405 23 7.6 (4.9 to 11.9)
Log rank P= 0.101
BC Specific Mortality (N0, Endocrine Therapy Alone)
RS < 18 632 7 1.9 (0.9 to 3.9)
RS 18-30 405 6 1.9 (0.9 to 4.4)
Log rank P= 0.681
Years

The box under each graph presents the number of patients at risk at each time point.
One-degree of freedom (df) log-rank P values were calculated from all data.

Multivariable Analyses

Table 2. Multivariable model of distant recurrence (n = 1245).

Variable	Comparison	Hazard ratio (95% CI)	*P*-value
Risk Group	18-30 vs <18	2.8 (1.5-5.1)	<0.001
	≥31 vs <18	6.0 (3.0-11.9)	
Age	50-69 vs <50	1.0 (0.5-1.9)	0.003
	≥70 vs <50	2.5 (1.2-5.3)	
Size	≥2 cm vs <2 cm	2.3 (1.4-3.7)	<0.001
Grade	2 vs 1	2.4 (0.9-6.6)	0.223
	3 vs 1	2.6 (0.9-7.6)	
Nodal status	N1mi vs N0	2.9 (1.7-5.2)	<0.001

A total of 295 patients were excluded from the analysis due to missing data.

Risk of Distant Recurrence in CT-treated and Untreated N0 Patients

- We analyzed the risk of distant recurrence in CT-treated vs untreated patients by TAILORx cut offs. Curves are not shown for RS<11 patients as none of them received CT, for RS 11-17 patients as <3% received CT, and for RS≥31 patients as <13% of them did not receive CT (**Figure 3**). It should be noted that patients were not randomized so there is a likely selection bias.

Figure 3. KM distant recurrence curves in N0 patients with RS 18-25 and 26-30 by CT use.

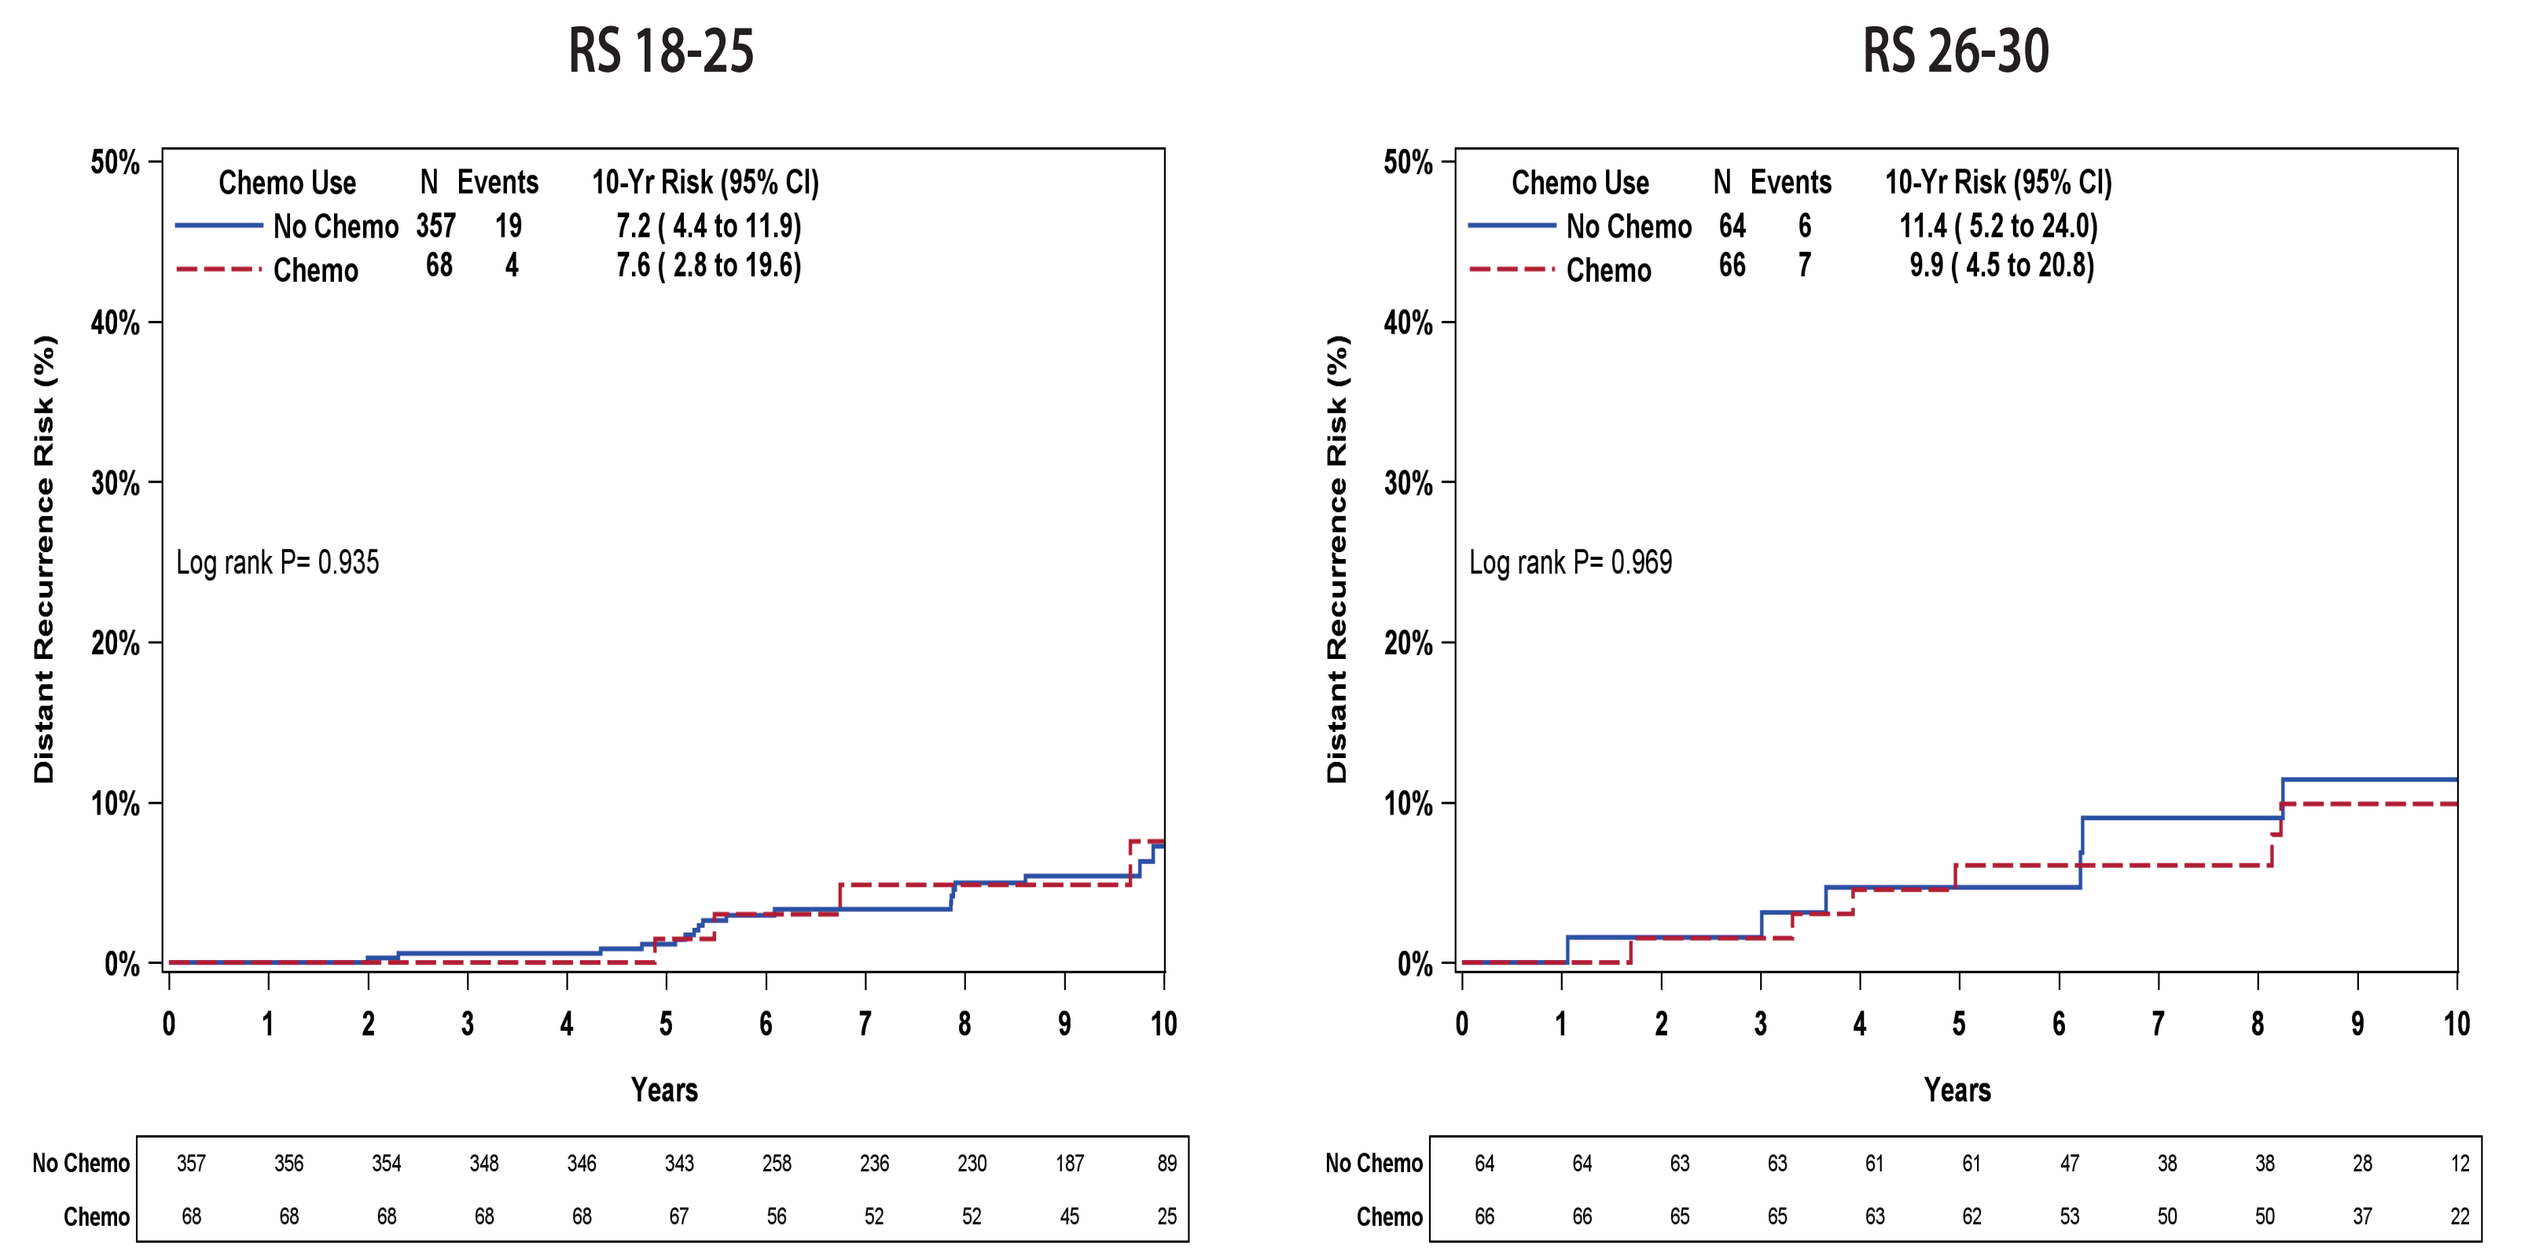
RS 18-25
Chemo Use N Events 10-Yr Risk (95% CI)
No Chemo 357 19 7.2 (4.4 to 11.9)
Chemo 68 4 7.6 (2.8 to 19.6)
Log rank P= 0.935
RS 26-30
No Chemo 64 6 11.4 (5.2 to 24.0)
Chemo 66 7 9.9 (4.5 to 20.8)
Log rank P= 0.969
Years

Log-rank P values were calculated from all data.

SUMMARY AND CONCLUSIONS

- These are the first reported 10-year outcome data from a large cohort of patients where the RS assay was included in adjuvant treatment decisions.
- This study is limited by its nonrandomized design with CT treatment greatly influenced by the RS result, the potential selection bias with respect to patients being tested with the RS assay, and the small sample sizes in some of the subgroup analyses.
- Nevertheless, the RS result was prognostic with respect to 10-year distant recurrence and 10-year BC death (P <0.001).
- The 10-year KM estimate of distant recurrence and BC death in N0 and N1mi patients with RS<18 was very low (N0, 4.0% and 1.8% respectively; N1mi, 1.0% and 1.7% respectively), despite low CT use in these patients (N0, 1.8%; N1, 6.1%). The distant recurrence and BC death rates in N0 RS<18 patients selected for endocrine therapy alone were 3.9% and 1.9%, respectively.
- Although the numbers are small and there was no randomization, little difference in 10 year outcomes between CT-treated and untreated patients were observed with RS<25. In the RS 18-25 group, 10-year risk of distant recurrence was 7.6% and 7.2% in CT-treated and untreated patients, respectively.
- These results strongly support that CT can be safely spared in patients with N0 and N1mi disease and RS<18 and suggest that the absolute CT benefit in N0 patients with RS 18-25 is unlikely.

REFERENCES

1. Paik S, et al. N Engl J Med. 2004;351:2817-2826. | 2. Toi M, et al. Cancer. 2010;116:3112-3118. | 3. Dowsett M, et al. J Clin Oncol. 2010;28:1829-1834. | 4. Sparano JA, et al. N Engl J Med. 2015;373:2005-2014. | 5. Paik S, et al. J Clin Oncol. 2006;24:3726-3734. | 6. Habel LA, et al. Breast Cancer Res. 2006;8:R25. | 7. Albain KS, et al. Lancet Oncol. 2010;11:55-65. | 8. Nitz U, et al. Breast Cancer Res Treat. 2017 [Epub ahead of print]. | 9. Gluz O, et al. J Clin Oncol. 2016;34:2341-9. | 10 Stemmer SM, et al. NPJ Breast Cancer. 2017;3:32. | 11. Stemmer SM, et al. NPJ Breast Cancer. 2017;3:332. | 12. Petkov VI, et al. NPJ Breast Cancer. 2016;2:16017. |
13. Senkus E, et al. Ann Oncol. 2015;26 Suppl 5:v8-30. | 14. NCCN clinical practice guidelines in oncology. Breast Cancer. Version 1.2017. Available at: http://www.nccn.org/professionals/physician_gls/pdf/breast.pdf. Accessed Oct 1, 2017. | 15. Coates AS, et al. Ann Oncol. Aug ;26:1533-1546. | 16. Harris LN, et al. J Clin Oncol. 2016;34:1134-1150. | 17. AJCC Cancer Staging Manual. 8th ed. 2017. Springer, New York. | 18. Pan H, et al. N Engl J Med. 2017;377:1836-1846. | 19. Early Breast Cancer Trialists' Collaborative Group, et al. Lancet. 2011;378:771-784. | 20. Burstein HJ, et al. J Clin Oncol. 2014;32:2255-2269.

Commercial relationships: This work was supported by grant 1132 from Teva Pharmaceuticals Ltd that was awarded to SM Stemmer via Mor Research Applications Ltd for the purpose of this study. SM Stemmer received grant funding from Teva, travel expenses from Genomic Health, Inc, and conducted research funded by Teva. D McCullough, C Svedman, and S Shak are em ployed by and have stock ownership in Genomic Health. L Soussan-Gutman is employed by Teva Pharmaceuticals. A Bareket-Samish is a paid consultant for Genomic Health and Teva Pharmaceuticals. N Ben-Baruch received honoraria from Teva, and served on the speaker's bureau of Genomic Health. O Rosengarten reports receiving payments for lectures and grants for traveling to conferences from Teva Pharmaceutical Industries Ltd. The remaining authors report no conflict of interest.

This presentation is the intellectual property of the author/presenter. Contact them at Stemmer@post.tau.ac.il for permission to reprint and/or distribute.